Pulmonary function in non-ambulatory patients with nmDMD from the STRIDE ataluren Registry and CINRG Duchenne Natural History Study: a matched cohort analysis

Andrés Nascimento Osorio,[1] Már Tulinius,[2] Filippo Buccella,[3] Isabelle Desguerre,[4] Janbernd Kirschner,[5] Eugenio Mercuri,[6] Joel Jiang,[7] Allan Kristensen,[7] Panayiota Trifillis,[7] Claudio L Santos[7] and Craig M McDonald,[8] on behalf of the STRIDE and CINRG DNHS investigators

[1]Hospital Sant Joan de Déu, Unidad de Patología Neuromuscular, Universidad de Barcelona, Barcelona, Spain; [2]Department of Pediatrics, Gothenburg University, Queen Silvia Children's Hospital, Gothenburg, Sweden; [3]Parent Project Italy APS, Rome, Italy; [4]APHP Necker – Enfants Malades Hospital, Paris V Descartes University, Neuromuscular Network FILNEMUS, Paris, France; [5]Department of Neuropediatrics, University Hospital Bonn, Bonn, Germany; [6]Department of Pediatric Neurology, Catholic University, Rome, Italy; [7]PTC Therapeutics Inc., South Plainfield, NJ, USA; [8]University of California Davis School of Medicine, Davis, CA, USA

1. Background

- Duchenne muscular dystrophy (DMD) is a severe, X-linked, recessive neuromuscular disorder affecting approximately 1 in every 3600–6000 live male births.[1–3]
- Owing to the absence of functional dystrophin protein, patients with DMD experience a progressive decline in functional ability, including loss of ambulation, and respiratory and cardiac complications, which lead to premature death.[1]
- In approximately 10–15% of patients, the disorder is caused by a nonsense mutation (nm) in the *DMD* gene, leading to a premature stop codon in the dystrophin mRNA, which prevents translation of a full-length, functional dystrophin protein.[4,5]
- Ataluren is an orally bioavailable drug that is designed to promote readthrough of the in-frame premature stop codon to produce full-length dystrophin.[6]
- Ataluren 40 mg/kg/day is indicated for the treatment of nmDMD in ambulatory patients aged ≥ 2 years[7] in member states of the European Union, and Iceland, Israel, Kazakhstan, Liechtenstein, Norway and the Republic of Korea, or aged ≥ 5 years in Brazil and Chile.[8]
 - Efficacy has not been demonstrated in non-ambulatory patients.
 - The presence of a nonsense mutation in the *DMD* gene should be determined by genetic testing.[7]
- The **S**trategic **T**argeting of **R**egistries and **I**nternational **D**atabase of **E**xcellence (STRIDE; ClinicalTrials.gov identifier: NCT02369731) is an ongoing, multicenter, observational registry providing real-world evidence regarding the use of ataluren in patients with nmDMD in routine clinical practice.
- The **C**ooperative **I**nternational **N**euromuscular **R**esearch **G**roup **D**uchenne **N**atural **H**istory **S**tudy (CINRG DNHS; ClinicalTrials.gov identifier: NCT00468832) was a prospective, longitudinal study of a total of 440 patients with DMD who were followed up between 2006 and 2016 at 20 centers worldwide as part of the academic clinical trial network, CINRG.[9,10]
- Here we examine whether, as of the latest data cut-off date of January 31, 2019, non-ambulatory STRIDE patients with nmDMD receiving ataluren plus standard of care (SoC; corticosteroid or palliative therapies) experienced a lesser decline in pulmonary function than non-ambulatory CINRG DNHS patients with DMD receiving SoC alone, using propensity-score matched analyses.

2. Methods

Study design

- The STRIDE Registry is a post-approval safety study of ataluren use in patients with nmDMD in routine clinical practice, requested by the Pharmacovigilance Risk Assessment Committee of the European Medicines Agency.
 - The study design is shown in **Figure 1**.
 - The registry can only be implemented in countries where ataluren is commercialized or available through an early access program (if applicable).
 - Patients will be followed up for ≥ 5 years, or until study withdrawal.
- Patients with DMD from the CINRG DNHS aged 2–28 years were enrolled at 20 centers in nine countries from 2006 to 2016.[9,10]
- Data from CINRG DNHS patients receiving SoC were used as a control to provide context for assessing the effects of ataluren plus SoC on patients in the STRIDE Registry.

Figure 1. STRIDE Registry post-approval safety study design.

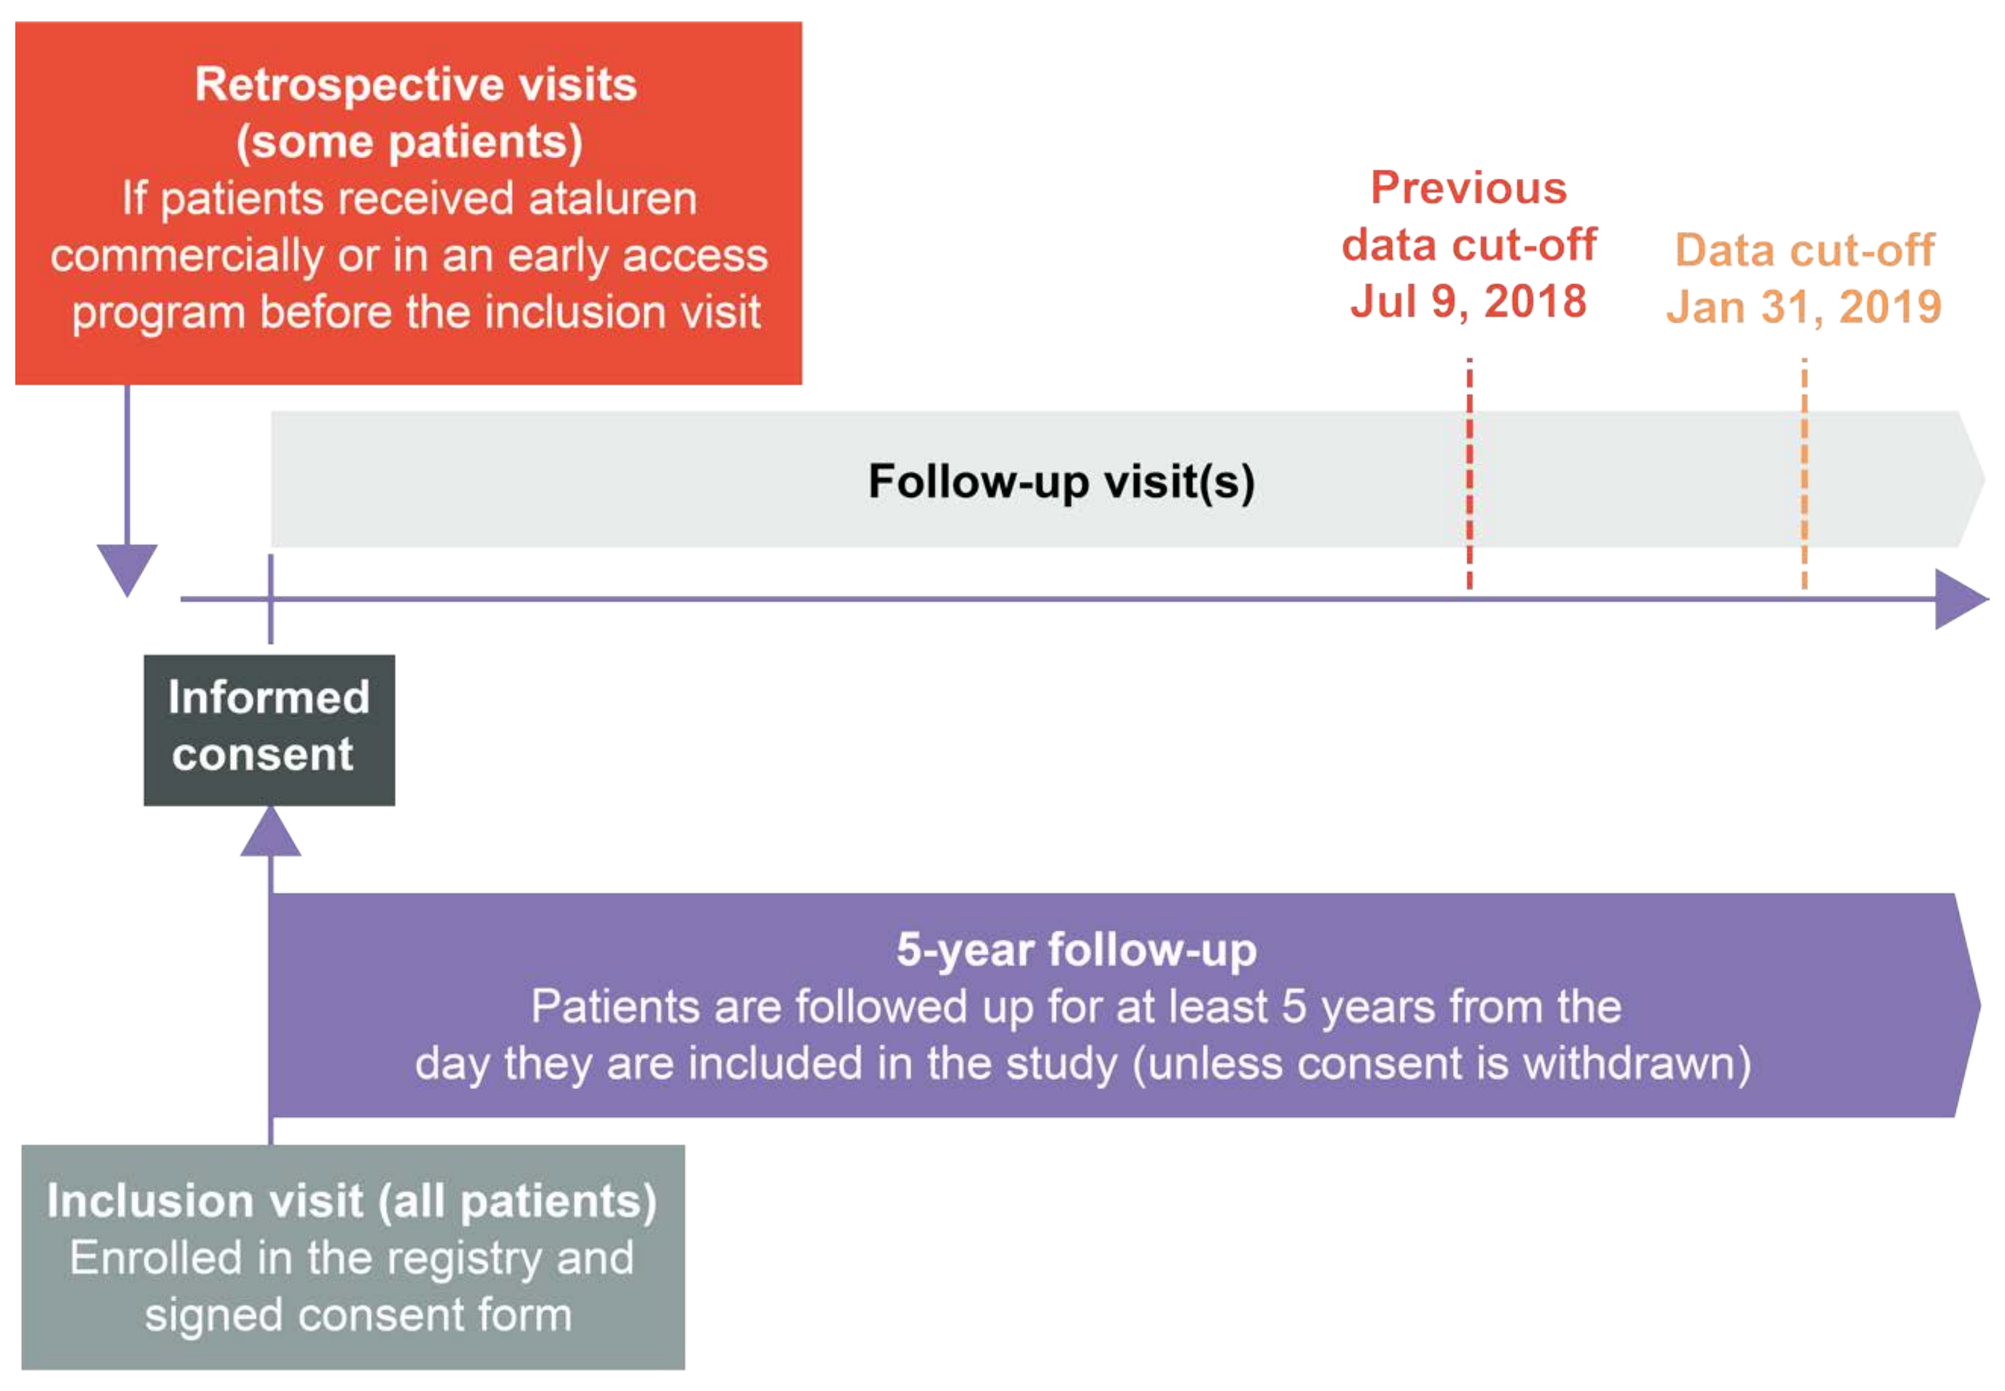
Retrospective visits (some patients)
If patients received ataluren commercially or in an early access program before the inclusion visit
Previous data cut-off Jul 9, 2018
Data cut-off Jan 31, 2019
Follow-up visit(s)
Informed consent
5-year follow-up
Patients are followed up for at least 5 years from the day they are included in the study (unless consent is withdrawn)
Inclusion visit (all patients)
Enrolled in the registry and signed consent form

STRIDE, Strategic Targeting of Registries and International Database of Excellence.

Study populations and data collection

STRIDE Registry

- Patients were eligible if they received ataluren as a commercial supply or as part of an early access program and provided written informed consent before participating in this study.
 - Dosage of ataluren received: 40 mg/kg/day (10, 10 and 20 mg/kg for morning, midday and evening doses, respectively).
- Patients were not eligible if they were receiving:
 - ataluren or placebo in an ongoing, blinded, randomized clinical trial
 - ataluren in any other ongoing clinical trial or early access program that prevented participation in this study.
- Data from enrolled patients were collected at the consent date; for patients who initiated ataluren as part of a commercial or early access program before enrollment, data for the period before enrollment were obtained retrospectively.

CINRG Duchenne Natural History Study

- Patients aged 2–4 years were included if they had a diagnosis of DMD confirmed by one or more of the following:
 - dystrophin immunofluorescence
 - immunoblotting
 - an out-of-frame deletion
 - *DMD* gene sequencing in the proband or sibling.
- Patients aged 5–28 years were included if they met the same criteria or had clinical symptoms of DMD and direct support for the diagnosis (a mutation in the *DMD* gene, abnormal dystrophin protein, or a combination of increased creatine kinase levels and an X-linked pedigree).[9,10]
- Patients were excluded from the present analysis if they had received investigational drugs in previous clinical trials.

Statistical analysis

- Propensity score matching was performed to identify non-ambulatory CINRG DNHS patients who were comparable to non-ambulatory STRIDE patients in the following covariates, which are established predictors of disease progression:[9,11,12]
 - age at onset of first symptoms
 - age at initiation of corticosteroid use
 - duration of deflazacort use
 - duration of use of other corticosteroids.
- To be eligible for propensity score matching, patients must have been non-ambulatory at the last assessment and have available data for age at loss of ambulation, lung function (forced vital capacity; FVC) and the covariates selected for the propensity-score matched analysis.
- Kaplan–Meier analyses were used to estimate the distribution of age at transition to the following endpoints ('events'): loss of ambulation; predicted FVC < 60%; predicted FVC < 50%; and FVC < 1 L. The distribution of these variables was compared between matched non-ambulatory STRIDE and CINRG DNHS patients using a log-rank test stratified by the duration of deflazacort and other corticosteroid use (< 1 month, ≥ 1 month and < 12 months, and ≥ 12 months).
- The hazard ratio (HR) of STRIDE Registry:CINRG DNHS and the corresponding 95% confidence intervals (CIs) were calculated using a Cox proportional hazard model stratified by durations of corticosteroid use, with study (STRIDE Registry or CINRG DNHS), age at first symptoms and age at initiation of corticosteroid use as covariates.

3. Results

STRIDE Registry patient disposition

- As of January 31, 2019, 224 patients with DMD had been enrolled in STRIDE; 220 male patients provided informed consent, did not fail screening and received at least one dose of ataluren (the evaluable/safety population) (**Figure 2**).
- Of 210 patients with genetically confirmed nmDMD in the effectiveness population, 22 were non-ambulatory (required full-time wheelchair use) at study entry and 24 lost ambulation during the study.

Figure 2. STRIDE Registry patient disposition.

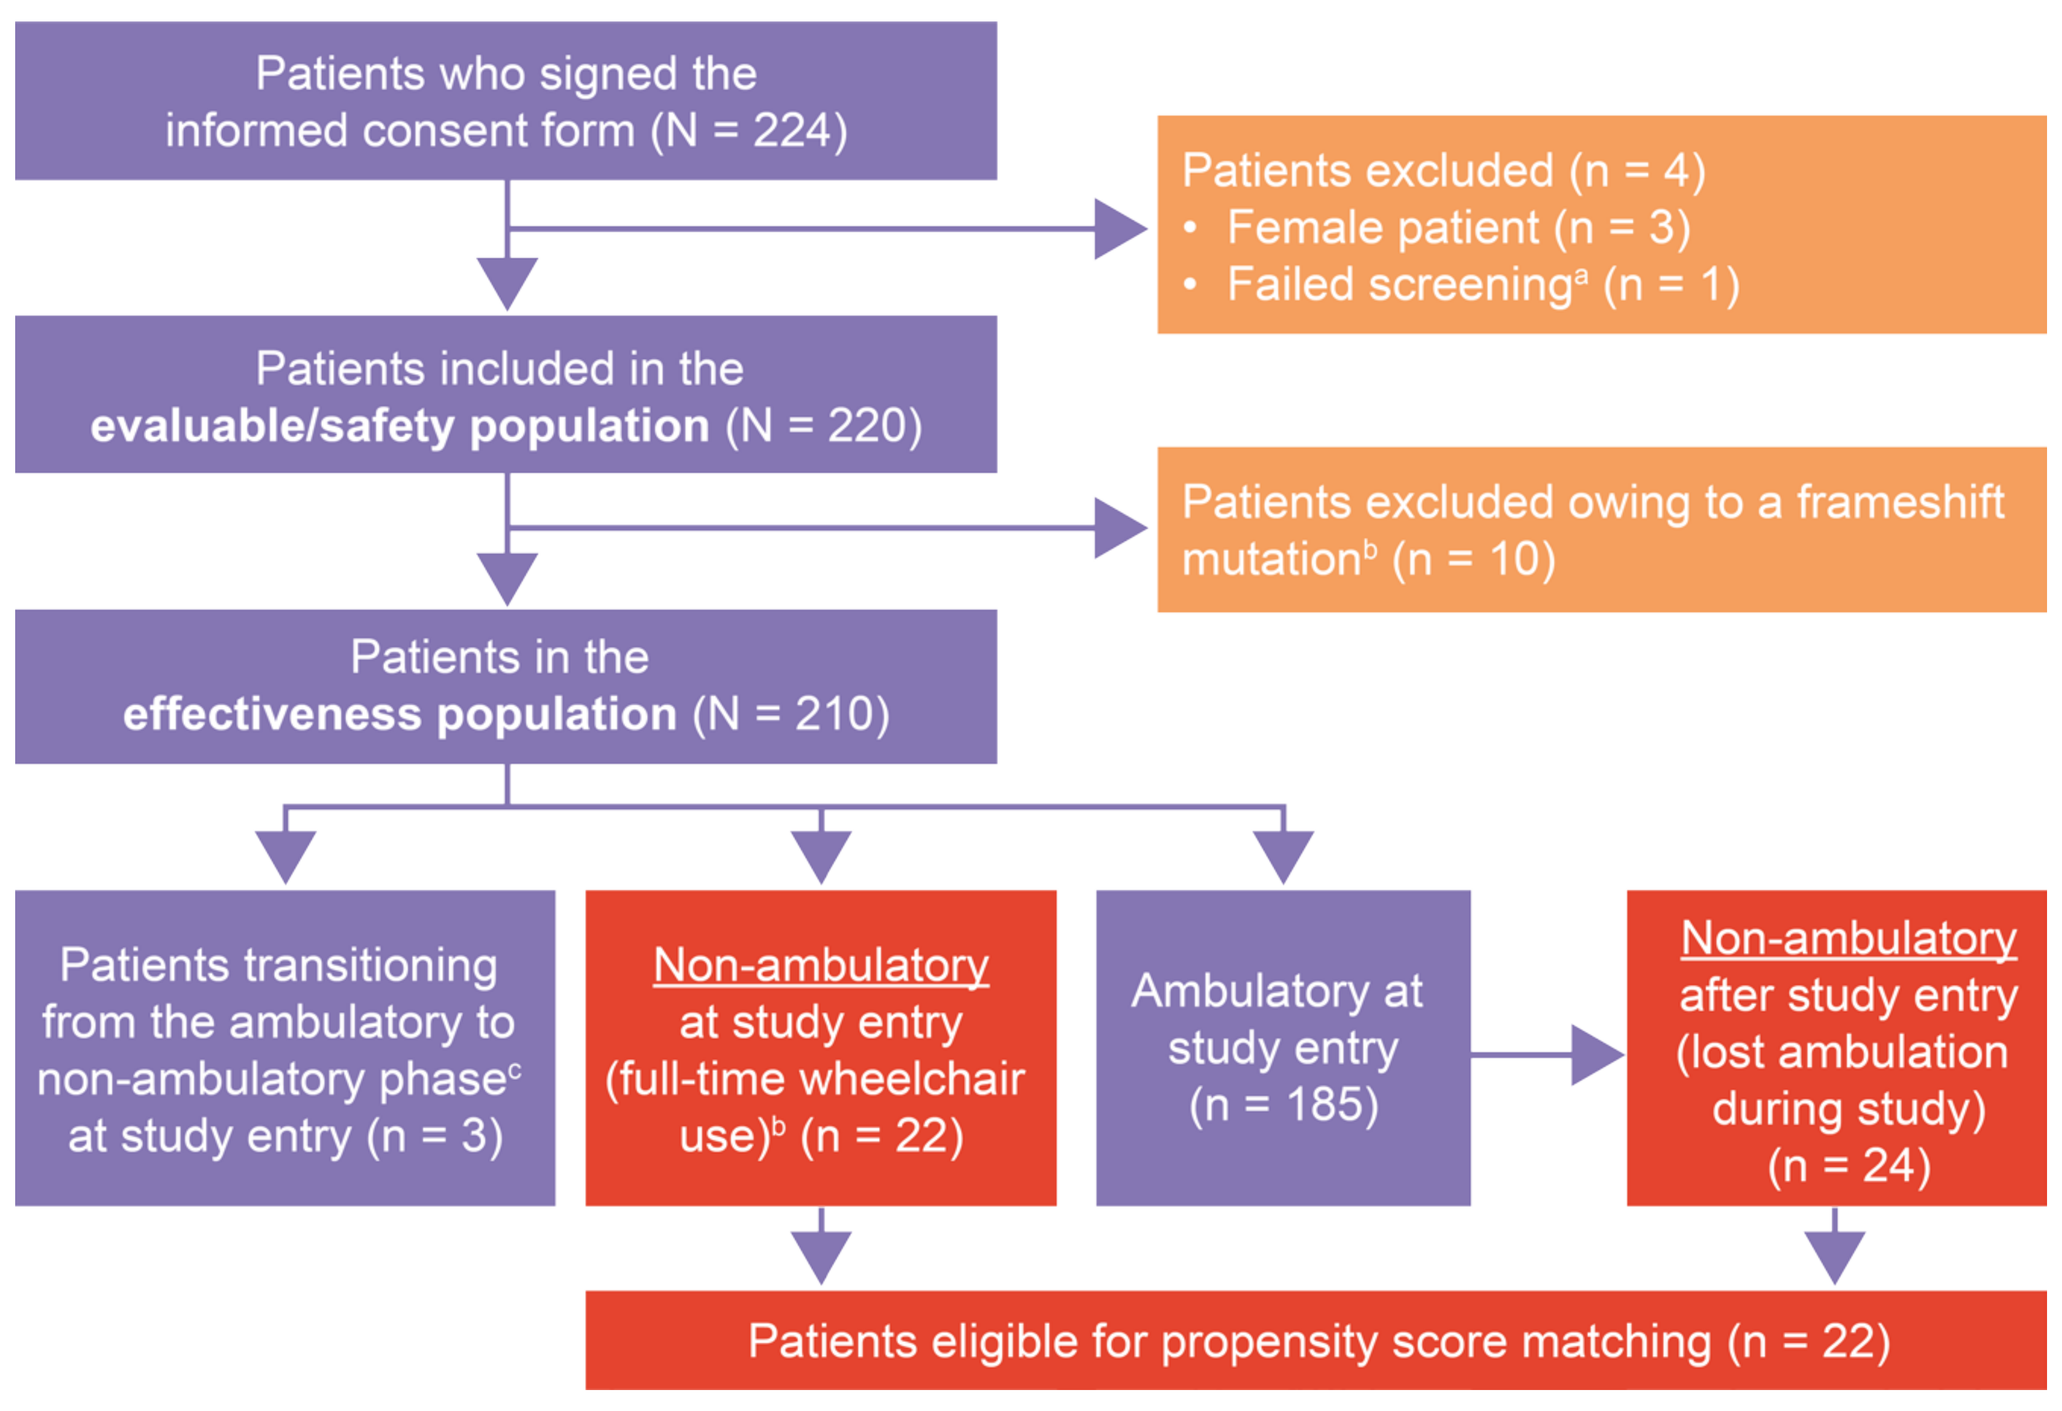
Patients who signed the informed consent form (N = 224)
Patients excluded (n = 4)
• Female patient (n = 3)
• Failed screening[a] (n = 1)
Patients included in the evaluable/safety population (N = 220)
Patients excluded owing to a frameshift mutation[b] (n = 10)
Patients in the effectiveness population (N = 210)
Patients transitioning from the ambulatory to non-ambulatory phase[c] at study entry (n = 3)
Non-ambulatory at study entry (full-time wheelchair use)[b] (n = 22)
Ambulatory at study entry (n = 185)
Non-ambulatory after study entry (lost ambulation during study) (n = 24)
Patients eligible for propensity score matching (n = 22)

[a]Screening failure due to a frameshift mutation.
[b]Ataluren is not indicated in non-ambulatory patients or patients with a frameshift mutation. The safety and efficacy of ataluren in non-ambulatory patients have not been established.
[c]Patients with a first 10-m run/walk test result of ≥ 30 seconds.
STRIDE, Strategic Targeting of Registries and International Database of Excellence.

Demographics and characteristics of propensity-score matched non-ambulatory STRIDE Registry and CINRG DNHS patients

- Of the 46 non-ambulatory STRIDE Registry patients, 22 were eligible for propensity score matching and therefore were matched to CINRG DNHS patients (N = 113), yielding a comparable population (N = 22) with respect to established predictors of disease progression (**Table 1**).[9,11,12]

Table 1. Demographics and characteristics of non-ambulatory patients in the STRIDE Registry and CINRG DNHS before and after propensity score matching.

Characteristic	Unmatched population		Propensity-matched population	
	STRIDE (N = 22)	CINRG DNHS (N = 113)	STRIDE (N = 22)	CINRG DNHS (N = 22)
Age at first symptoms, years				
Mean (SD)	3.0 (2.0)	3.4 (1.8)	3.0 (2.0)	3.4 (2.2)
p value	0.3969		0.5104	
Age at first corticosteroid use (excluding treatment-naive patients),[a] years	n = 19	n = 98	n = 19	n = 19
Mean (SD)	7.6 (2.3)	7.3 (1.9)	7.6 (2.3)	7.7 (2.4)
p value	0.5879		0.8920	
Deflazacort duration,[b] n (%)				
< 1 month or treatment-naive	18 (81.8)	72 (63.7)	18 (81.8)	19 (86.4)
≥ 1 to < 12 months	0 (0.0)	3 (2.7)	0 (0.0)	0 (0.0)
≥ 12 months	4 (18.2)	38 (33.6)	4 (18.2)	3 (13.6)
p value	0.2344		0.6802	
Other corticosteroid duration,[b] n (%)				
< 1 month or treatment-naive	6 (27.3)	50 (44.3)	6 (27.3)	5 (22.7)
≥ 1 to < 12 months	0 (0.0)	10 (8.9)	0 (0.0)	0 (0.0)
≥ 12 months	16 (72.7)	53 (46.9)	16 (72.7)	17 (77.3)
p value	0.0599		0.7277	

Propensity score model covariates include age at first symptoms, age at first corticosteroid use, duration of deflazacort use and duration of use of corticosteroids other than deflazacort.
[a]Treatment-naive patients were excluded to calculate the true age at first corticosteroid use.
[b]Corticosteroid duration is calculated from the date at which corticosteroid use was started to the date of loss of ambulation or the date of the last assessment.
CINRG DNHS, Cooperative International Neuromuscular Research Group Duchenne Natural History Study; SD, standard deviation; STRIDE, Strategic Targeting of Registries and International Database of Excellence.

Age at loss of ambulation and pulmonary function tests for propensity-matched non-ambulatory patients

- The median ages at loss of ambulation for STRIDE Registry and CINRG DNHS patients (each N = 22) were 12.4 years and 11.1 years, respectively (**Figure 3a**).
 - While enrolled in the registry, STRIDE patients received ataluren for a mean (95% CI) of 302 (163, 440) days before loss of ambulation.
- In the STRIDE Registry population, 18.2% (4/22) had a predicted FVC of < 60%, whereas in the matched CINRG DNHS population, 68.2% (15/22) reached this endpoint. The median age at predicted FVC < 60% was significantly higher for patients in the STRIDE Registry than for patients in the matched CINRG DNHS population (HR, 0.150; $p < 0.01$; **Figure 3b**).
 - STRIDE patients received ataluren for a mean (95% CI) of 661 (495, 826) days before reaching the event predicted FVC < 60%, or being censored.
- None of the 22 STRIDE patients had a predicted FVC of < 50%, whereas 54.5% of patients (12/22) in the matched CINRG DNHS population reached this endpoint (**Figure 3c**).
- Similarly, none of the 22 STRIDE patients had an FVC < 1 L, whereas 22.7% of patients (5/22) in the matched CINRG DNHS population reached this endpoint (**Figure 3d**).

3. Results (continued)

Figure 3. Age at a) loss of ambulation, b) predicted FVC < 60%, c) predicted FVC < 50% and d) FVC < 1 L for propensity-score matched patients from the STRIDE Registry and CINRG DNHS.

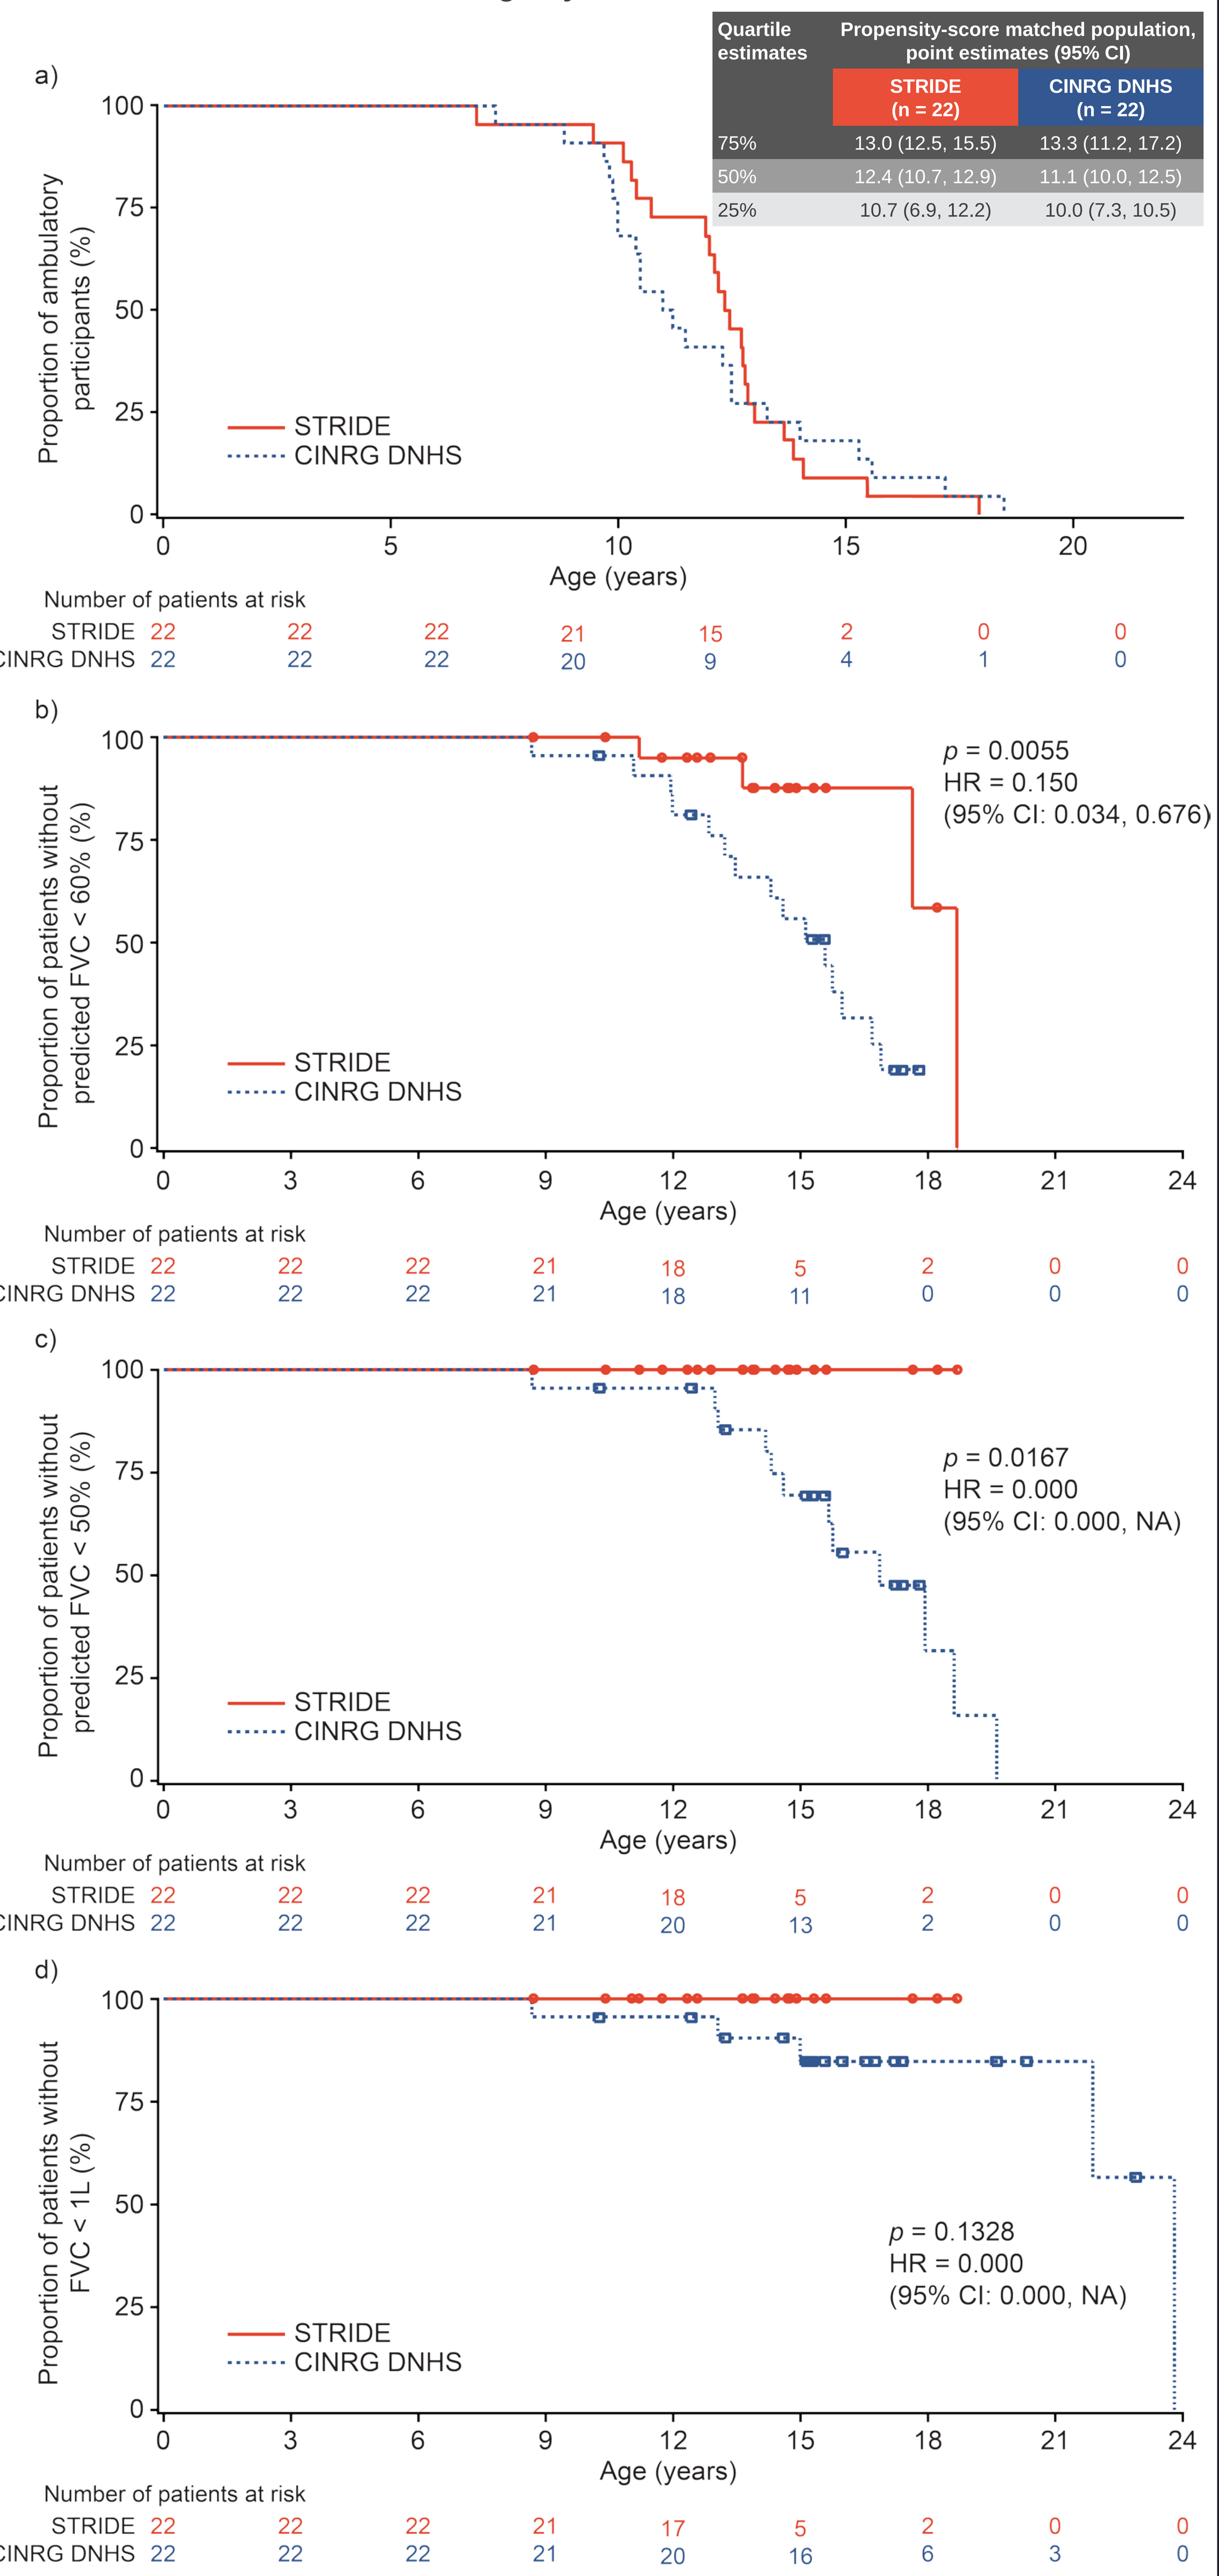
a)
Proportion of ambulatory participants (%)
Age (years)
STRIDE
CINRG DNHS
Number of patients at risk
STRIDE 22 22 22 21 15 2 0 0
CINRG DNHS 22 22 22 20 9 4 1 0
b)
Proportion of patients without predicted FVC < 60% (%)
p = 0.0055
HR = 0.150
(95% CI: 0.034, 0.676)
Age (years)
Number of patients at risk
STRIDE 22 22 22 21 18 5 2 0 0
CINRG DNHS 22 22 22 21 18 11 0 0 0
c)
Proportion of patients without predicted FVC < 50% (%)
p = 0.0167
HR = 0.000
(95% CI: 0.000, NA)
Age (years)
Number of patients at risk
STRIDE 22 22 22 21 18 5 2 0 0
CINRG DNHS 22 22 22 21 20 13 2 0 0
d)
Proportion of patients without FVC < 1L (%)
p = 0.1328
HR = 0.000
(95% CI: 0.000, NA)
Age (years)
Number of patients at risk
STRIDE 22 22 22 21 17 5 2 0 0
CINRG DNHS 22 22 22 21 20 16 6 3 0

Quartile estimates	Propensity-score matched population, point estimates (95% CI)	
	STRIDE (n = 22)	CINRG DNHS (n = 22)
75%	13.0 (12.5, 15.5)	13.3 (11.2, 17.2)
50%	12.4 (10.7, 12.9)	11.1 (10.0, 12.5)
25%	10.7 (6.9, 12.2)	10.0 (7.3, 10.5)

	Predicted FVC < 60 %		Predicted FVC < 50 %		FVC < 1 L	
	STRIDE	CINRG DNHS	STRIDE	CINRG DNHS	STRIDE	CINRG DNHS
Patients, n (%)						
Assessed	22	22	22	22	22	22
With events,[a] n (%)	4 (18.2)	15 (68.2)	0 (0.0)	12 (54.5)	0 (0.0)	5 (22.7)
Censored	18 (81.8)	7 (31.8)	22 (100.0)	10 (45.5)	22 (100.0)	17 (77.3)
Age at event, years						
Median (95% CI)	18.7 (17.7, 18.7)	15.6 (13.2, 16.7)	NA (NA, NA)	16.9 (14.3, 18.6)	NA (NA, NA)	23.8 (21.9, 23.8)
Min, max[b]	8.7+, 18.7	8.7, 17.8+	8.7+, 18.7	8.7, 19.6	8.7+, 18.7+	8.7, 23.8
p value[c]	0.0055		0.0167		0.1328	
HR (95% CI)[d]	0.150 (0.034, 0.676)		0.000 (0.000, NA)		0.000 (0.000, NA)	

Propensity score model covariates include age at first symptoms, age at first corticosteroid use, duration of deflazacort use, and duration of use of corticosteroids other than deflazacort. Censor dates for censored patients in the STRIDE Registry were derived from the last assessment date across treatment, physical examination, vital signs, 6-minute walk distance, timed function tests, North Star Ambulatory Assessments, percentage of predicted FVC and upper limb function tests. Corticosteroid duration is calculated from the date at which corticosteroid use was started to the date of loss of ambulation or the date of the last assessment.
[a]Event = loss of ambulation, predicted FVC < 60%, predicted FVC < 50% or FVC < 1 L.
[b]'+' indicates censored observation.
[c]*p* value is from a log-rank test stratified by deflazacort and other corticosteroid usage durations.
[d]The HR is from stratified (by durations of deflazacort and other corticosteroid use) Cox regression with study (STRIDE Registry or CINRG DNHS), age at first symptoms and age at first corticosteroid use as covariates. The HR is STRIDE Registry:CINRG DNHS.
CI, confidence interval; CINRG DNHS, Cooperative International Neuromuscular Research Group Duchenne Natural History Study; FVC, forced vital capacity; HR, hazard ratio; max, maximum; min, minimum; NA, not available; STRIDE, Strategic Targeting of Registries and International Database of Excellence.

4. CONCLUSIONS

- In propensity-score matched analyses of non-ambulatory patients from the STRIDE Registry and CINRG DNHS, STRIDE patients received ataluren for a mean of 302 days before loss of ambulation, which occurred ~1.3 years later than for CINRG DNHS patients (not statistically significant).
- The same cohort of STRIDE patients received ataluren for a mean of 661 days before decline of pulmonary function to a predicted FVC < 60%, which occurred at a statistically significantly higher median age for STRIDE patients (18.7 years) than for CINRG DNHS patients (15.6 years), or being censored.
- There was a trend toward a delay in the age at decline in pulmonary function in STRIDE patients compared with CINRG DNHS patients, as measured by predicted FVC < 50% and FVC < 1 L.
- These data suggest that treatment with ataluren in addition to SoC may delay loss of ambulation, and in turn, pulmonary functional decline, in patients with nmDMD.
- Future comparisons of data from the STRIDE Registry with CINRG DNHS data will provide further real-world insights into the long-term effectiveness of ataluren for the treatment of patients with nmDMD.

References
1. Birnkrant DJ *et al. Lancet Neurology* 2018;17:251–67.
2. Bushby K *et al. Lancet Neurol* 2010;9:77–93.
3. Ellis JA *et al. Neuromuscul Disord* 2013;23:682–9.
4. Bello L *et al. Acta Myologica* 2016;35:122–7.
5. Pichavant C *et al. Mol Ther* 2011;19:830–40.
6. Roy B *et al. Proc Natl Acad Sci USA* 2016;113:12508–13.
7. PTC Therapeutics International Ltd. Summary of product characteristics, Translarna. European Medicines Agency, 2018. Available from: www.ema.europa.eu/documents/product-information/translarna-epar-product-information_en.pdf (Accessed January 9, 2020).
8. Data on file.
9. McDonald CM *et al. Lancet* 2018;391:451–61.
10. McDonald CM *et al. Muscle Nerve* 2013;48:357–68.
11. Ricotti V *et al. J Neurol Neurosurg Psychiatry* 2013;84:698–705.
12. Ciafaloni E *et al. J Pediatr Rehabil Med* 2016;9:5–11.

Disclosures
The STRIDE Registry is sponsored by PTC Therapeutics. Medical writing support was provided by Tim Ellison, PhD, of PharmaGenesis London, London, UK, and was funded by PTC Therapeutics. **ANO** has received speaker and consultancy fees from Biogen, PTC Therapeutics and Sarepta Therapeutics, and is an investigator on clinical trials sponsored by Biogen, F. Hoffmann-La Roche, Italfarmaco, Sarepta Therapeutics and TAMDMD. **MT** has received lecture fees from Biogen and PTC Therapeutics, has acted as a consultant on DMD clinical trials for BioMarin, Catabasis Pharmaceuticals, PTC Therapeutics, ReveraGen BioPharma and Sarepta Therapeutics, and as an advisory board member for AveXis, Biogen, PTC Therapeutics and Sarepta Therapeutics. **FB** has received consultancy fees from Pfizer, PTC Therapeutics, Santhera Pharmaceuticals and Sarepta Therapeutics. **ID** has received consultancy fees from AveXis, Biogen, BioMarin and PTC Therapeutics. **JK** has acted as a consultant for AveXis, Biogen, PTC Therapeutics, Roche and Sarepta Therapeutics, and has received research support for taking part in clinical research from AveXis, Biogen, Novartis, PTC Therapeutics, Roche and Santhera Pharmaceuticals. **EM** has acted as an advisory board member for AveXis, Biogen, BioMarin, Bristol-Myers Squibb, Ionis Pharmaceuticals, Italfarmaco, Prosensa, PTC Therapeutics, Roche, Santhera Pharmaceuticals, Sarepta Therapeutics and Summit Therapeutics. **JJ**, **AK**, **PT** and **CLS** are employees of PTC Therapeutics, Inc. **CMM** has acted as a consultant on clinical trials of DMD for Astellas, BioMarin, Capricor Therapeutics, Cardero Therapeutics, Catabasis Pharmaceuticals, Eli Lilly, FibroGen, Italfarmaco, Pfizer, PTC Therapeutics, Santhera Pharmaceuticals and Sarepta Therapeutics, and has received research support for clinical trials from BioMarin, Capricor Therapeutics, Eli Lilly, Italfarmaco, Pfizer, PTC Therapeutics, Roche, Santhera Pharmaceuticals and Sarepta Therapeutics.

Acknowledgments
We thank the patients and their families for their participation in this study, and individuals involved in the conduct of this study and the collection of data, particularly the STRIDE principal investigators and study coordinators.

The CINRG DNHS was funded by grants from the US Department of Education/NIDRR (#H133B031118, #H133B090001), the US Department of Defense (#W81XWH-09-1-0592), the National Institutes of Health (#UL1RR031988,#U54HD053177, #UL1RR024992, #U54RR026139, #2U54HD053177, #G12RR003051, #1R01AR061875, #RO1AR062380) and Parent Project Muscular Dystrophy. We thank the CINRG DNHS study participants as well as the members of the participating CINRG DNHS sites and central team.

CINRG DNHS investigators (sorted by country): Argentina: A Dubrovsky (Buenos Aires). Australia: A Kornberg, M Ryan (Melbourne, VIC); R Webster (Sydney, NSW). Canada: JK Mah (Calgary, AB); WD Biggar, H Kolski (Edmonton, AB); LC McAdam (Toronto, ON). India: S Chidambaranathan, V Vishwanathan (Chennai). Israel: Y Nevo (Jerusalem). Italy: K Gorni (Milan). Puerto Rico: J Carlo (San Juan). Sweden: M Tulinius (Gothenburg). USA: T Lotze (Houston, TX); TE Bertorini (Memphis, TN); JW Day, P Karachunski (Minneapolis, MN); H Abdel-Hamid, PR Clemens (Pittsburgh, PA); J Teasley (Richmond, VA); JB Bodensteiner, S Driscoll, N Kuntz (Rochester, MN); RT Abresch, EK Henricson, NC Joyce, CM McDonald (Sacramento, CA); AM Connolly, A Pestronk (St Louis, MO); A Cnaan, T Duong, R Leshner, LP Morgenroth, C Tesi-Rocha, M Thangarajh (Washington DC).

Scan QR code for poster

PRESENTED AT PARENT PROJECT MUSCULAR DYSTROPHY 22-25 SEPTEMBER, 2020

COPYRIGHT © 2020 PTC THERAPEUTICS INC., ALL RIGHTS RESERVED
TRANSLARNA™ IS A TRADEMARK OF PTC THERAPEUTICS, INC.